POPULATION PHARMACOKINETIC META ANALYSIS: INHIBITION BY QUINIDINE OF THE FIRST-PASS AND SYSTEMIC METABOLISM OF DEXTROMETHORPHAN TO DEXTRORPHAN

J. Grenier(1) Ph.D., JR. Lavigne(1) M.Sc., LE. Pope(2) Ph.D.

(1) Clinical Pharmacology Department, Celerion, Montréal, Qc, Canada
(2) Avanir Pharmaceuticals, Aliso Viejo, CA, USA

INTRODUCTION

Zenvia is a combination of 2 approved drugs, Dextromethorphan (DM) and Quinidine (Q), and is being developed for the treatment of Pseudobulbar Affect (PBA). The limited systemic delivery to the central nervous system (CNS) of DM may be a limiting factor of its efficacy in the treatment of different neurological disorders. Q is used as an inhibitor of DM metabolism by CYP2D6 enzymes to increase its bioavailability. A dose combination of 30 mg DM with 30 mg Q b.i.d. was used during the early development of Zenvia. In order to improve the safety profile of the drug, the dose of Q was subsequently reduced to 10 mg b.i.d. The current dose formulations of Zenvia in development for the treatment of PBA are DM 20 mg/Q 10 mg, and DM 30 mg/Q 10 mg. This dose of Q in Zenvia is 1-3% of that used to treat arrhythmias.

OBJECTIVE

The objectives of this study were to determine the population pharmacokinetic (PK) parameters of Q, DM and its metabolite Dextrorphan (DX) in plasma after single and multiple doses of Zenvia and to identify the impact of demographic covariates on the population PK parameter estimates.

DATA

The results of a series of Phase I - III studies were combined in order to perform a population PK analysis of Q, DM and its metabolite DX in plasma.

Phase I: double-blind, randomized, placebo-controlled study in healthy volunteers.

- Dosing regimens (for 8 days): 45/30 mg DM/Q b.i.d., 30/10 mg DM/Q b.i.d., 30/10 mg DM/Q t.i.d., 60/15 mg DM/Q b.i.d., 60/15 mg DM/Q once daily and placebo b.i.d.
- Plasma samples on Day 1 and Day 8.
- 60 subjects receiving active treatment were included.
- All subjects had extensive metabolizer (EM) genotype for CYP2D6.

Phase I: thorough QT trial (50 healthy volunteers)

- Clinical dose of 30 mg DM and 10 mg Q b.i.d.
- Plasma samples on Day 4.
- All subjects had the EM genotype for CYP2D6.

Phase III: multicenter safety and efficacy study in patients

- Dosing regimens were 30/10 mg DM/Q b.i.d. and 20/10 mg DM/Q b.i.d.
- 24 of the 326 patients had a rich PK sampling schedule at steady-state on Day 29.
- 22 were EM, 1 was ultra-metabolizer (UM) and 1 was intermediate-metabolizer (IM) of CYP2D6.

Data from 133 subjects were analyzed using a total of 5730 data points for Q, DM and DX combined. A set of 70 subjects from other studies were used for the external validation.

METHODOLOGY

Sequential modeling:

1. Q data were fitted alone.
2. Q PK parameters were then fixed for each individual.
3. DM and DX plasma data were fitted using predicted Q concentrations as input for the metabolic inhibition by Q of the metabolic conversion of DM to DX.

Covariates:

Age, gender, race, height, weight, body mass index - impact assessed graphically.

Internal and external validation:

Standardized visual predictive checks (SVPC) were evaluated for the internal and external validation of the final population PK models.[1]

Software: ADAPT 5, Version 5.0.34, August 25, 2009, using ITS algorithm.[2]

RESULTS AND DISCUSSION

The population PK model that best described the PK of Q in plasma was a 2-compartment model with linear absorption and elimination and a lag time for absorption. The population PK model that best described the PK of DM and DX in plasma was a model with 2 first-order constants of absorption with lag times, 2 compartments for DM and 1 compartment for DX. The metabolic conversion of DM to DX was described by a sigmoidal inhibition model related to Q concentrations, both at first-pass and systemically.

Figure 1: Structural PK Model for Q, DM and DX in Plasma

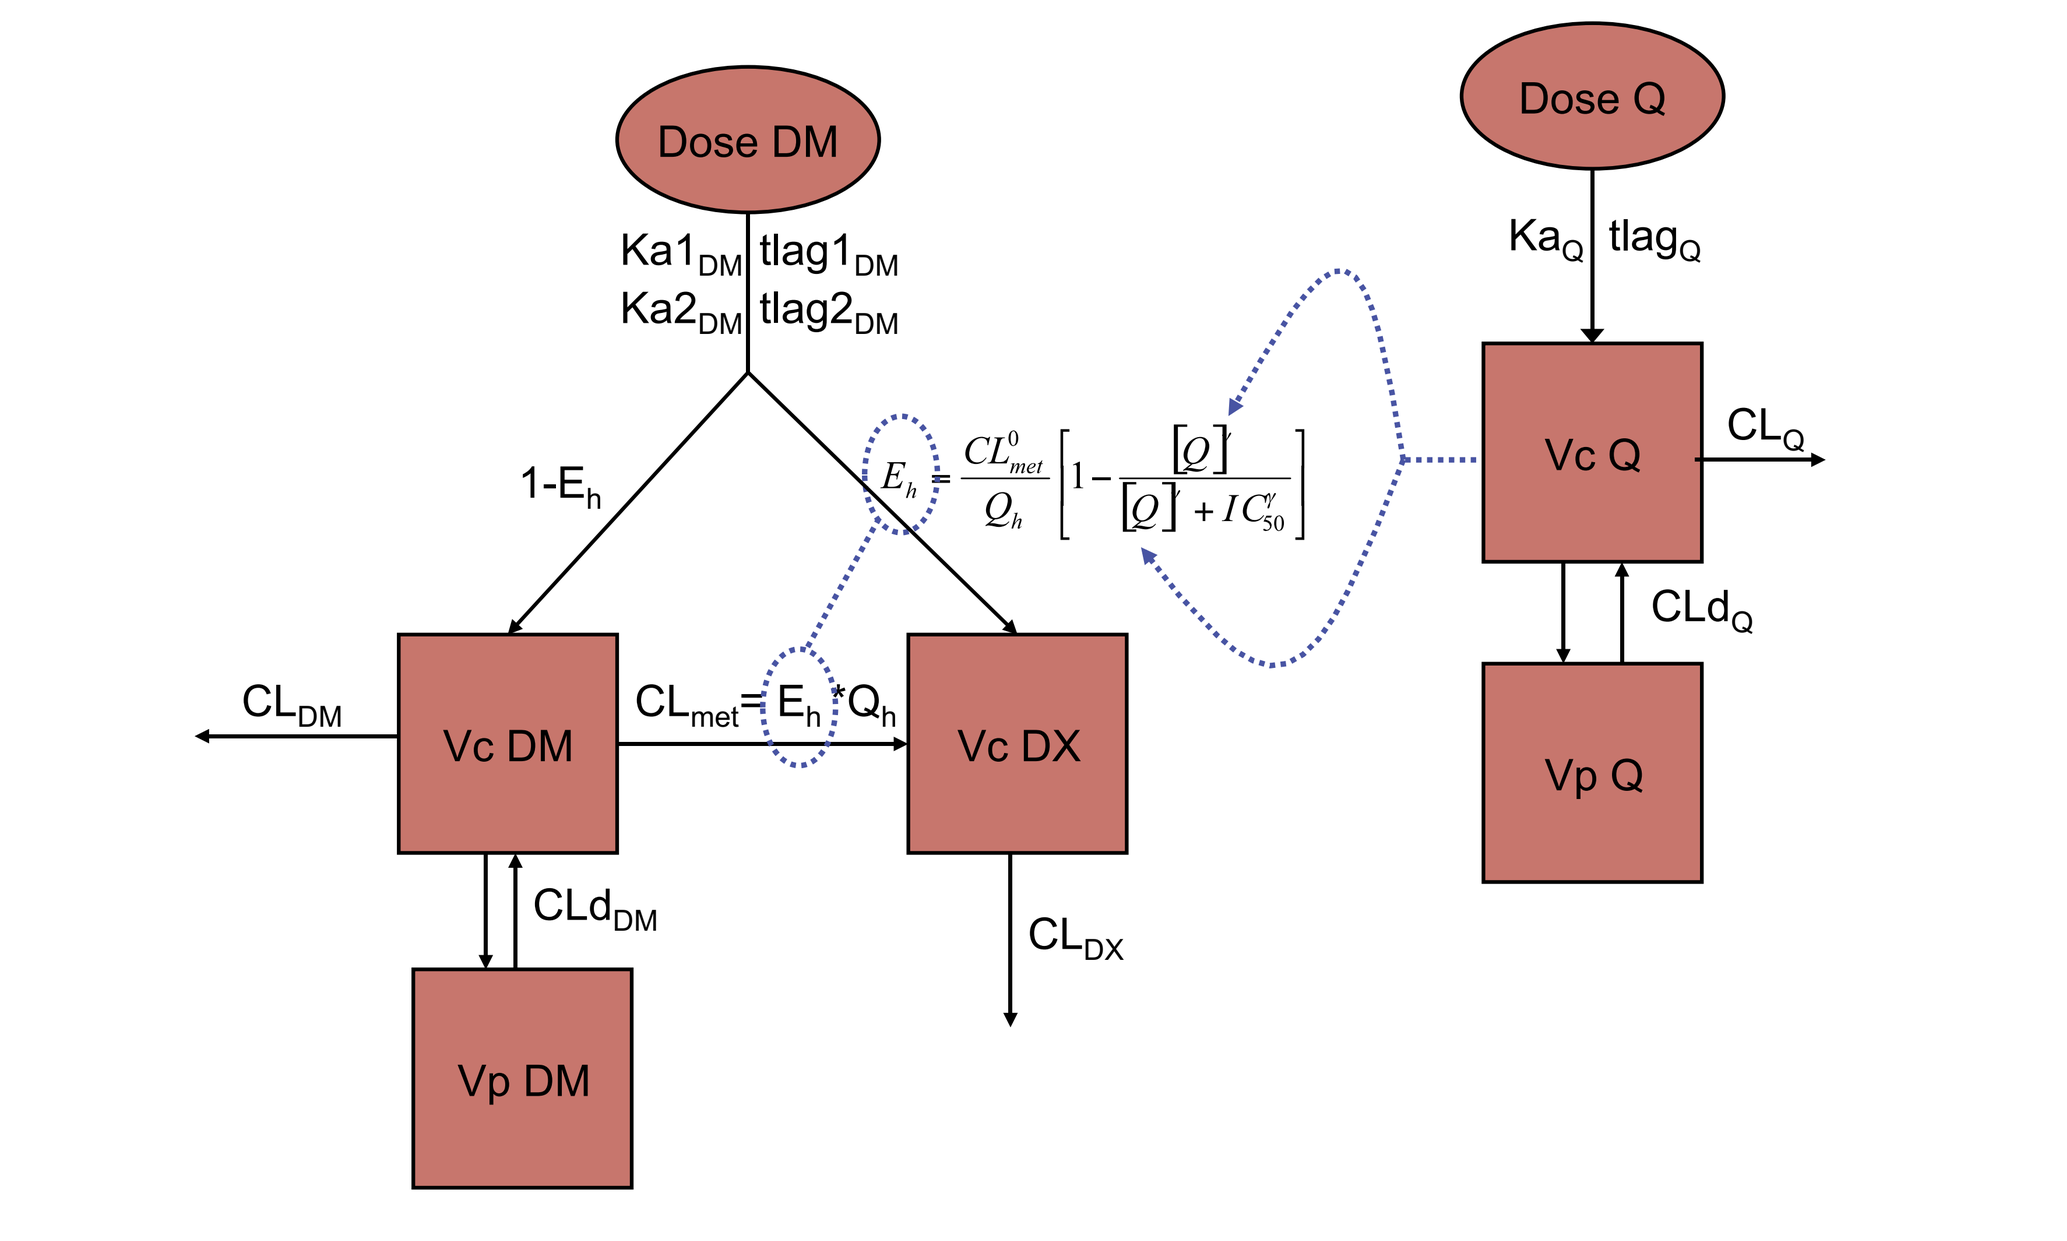
Dose DM
$Ka1_{DM}$ $tlag1_{DM}$
$Ka2_{DM}$ $tlag2_{DM}$
$1-E_h$
$E_h = \frac{CL^0_{met}}{Q_h}\left[1-\frac{[Q]}{[Q]+IC_{50}^{\gamma}}\right]$
CL_{DM}
Vc DM
$CL_{met} = E_h * Q_h$
Vc DX
CLd_{DM}
Vp DM
CL_{DX}
Dose Q
Ka_Q $tlag_Q$
Vc Q
CL_Q
CLd_Q
Vp Q

Ka=first-order constant of absorption; tlag=absorption lag time; Eh=hepatic extraction ratio (constrained between 0 and 1); Qh=hepatic blood flow; CL=systemic clearance; CLd=distribution clearance; Vc=volume of central compartment; Vp=volume of peripheral compartment; CL0met= metabolic clearance of DM to DX in the absence of Q; CLmet= metabolic clearance of DM to DX; IC50=quinidine concentration at half the maximum inhibition effect; γ =Hill coefficient

Table 1: Population PK Parameters for Quinidine

Parameters	Mean	Inter-subject variability (%CV)
tlag (h)	0.357	101
Ka (h^{-1})	2.32	38.9
CL/F (L/h)	20.7	33.1
Vc/F (L)	173	30.1
CLd (L/h)	3.02	64.9
Vp (L)	33.3	38.7
Intra-subject variability (%CV)	5.81	

Table 2: Population PK Parameters for DM and DX

	Parameters	Mean	Inter-subject variability (%CV)
Absorption	tlag1 (h)	0.597	38.9
	Ka1 (h^{-1})	0.791	57.7
	tlag2 (h)	1.38	22.2
	Ka2 (h^{-1})	0.555	47.6
	prop1*	1.20	72.8
DM	CL/F (L/h)	11.7	43.2
	Vc/F (L)	431	31.1
	CLd/F (L/h)	66.4	55.8
	Vp/F (L)	409	32.0
DX	CL/F (L/h)	9.59	14.3
	Vc/F (L)	17.4	31.7
Metabolism	CL^0_{met} (L/H)	57.5	37.4
	IC50 (mcg/L)	19.3	80.7
	γ	1.21	58.7
DM - Intra-subject variability (%CV)		5.26	
DX - Intra-subject variability (%CV)		4.24	

*Proportion of the dose being absorption by the Ka1 path = 1/(1+prop1)

The residual error model was proportional and additive and parameters were normally distributed in the final model presented in Figure 1. The key feature of this model is the inhibition of both first-pass and systemic metabolism.

Figure 2: Typical Plasma Concentration vs Time Profiles

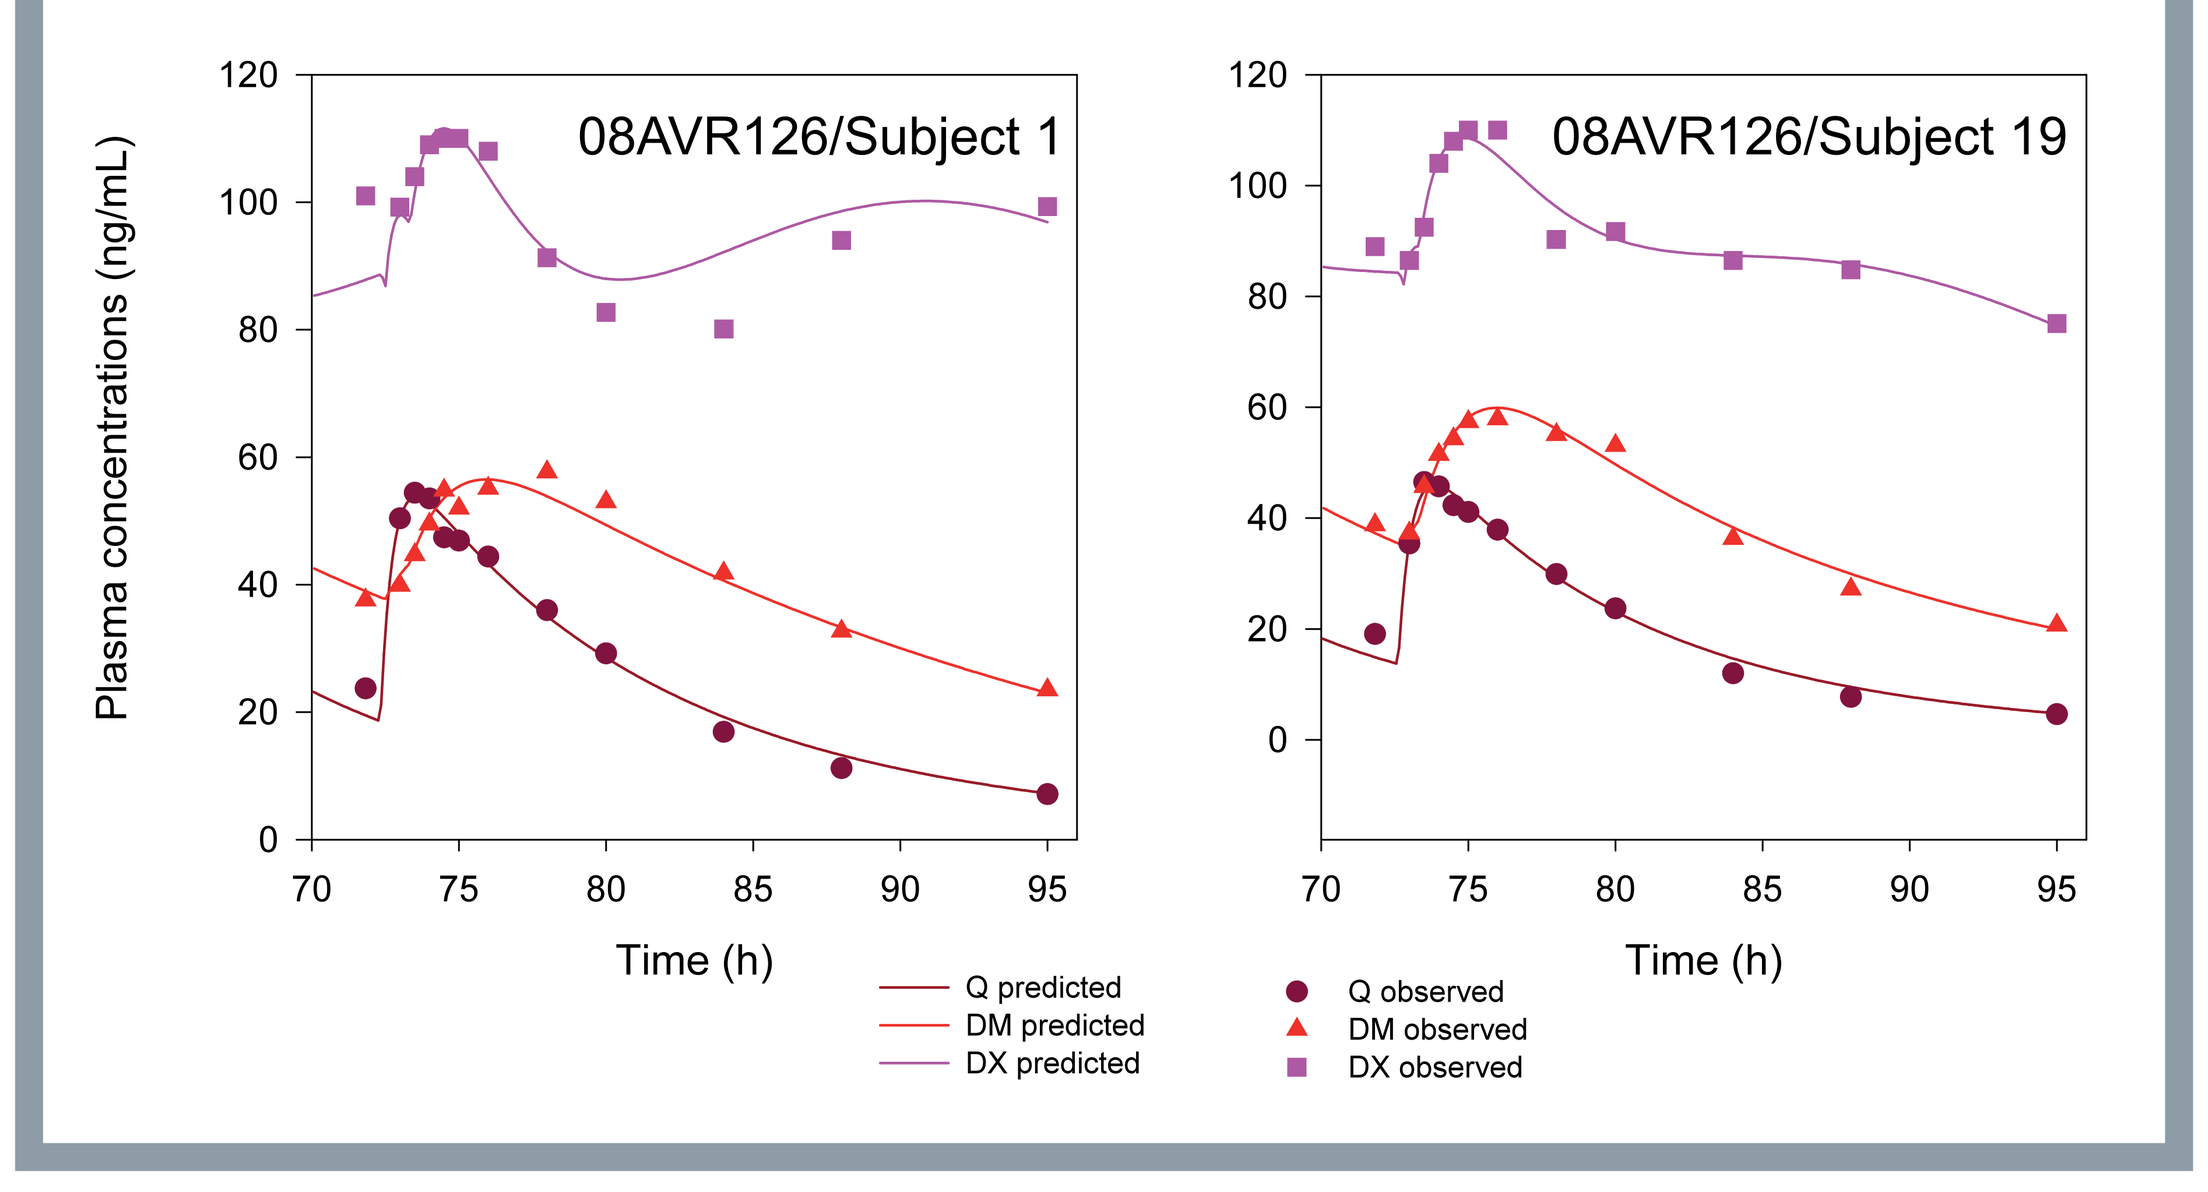
08AVR126/Subject 1
08AVR126/Subject 19
Plasma concentrations (ng/mL)
Time (h)
Q predicted
DM predicted
DX predicted
Q observed
DM observed
DX observed

The model discrimination and selection for DM/DX was based on data from EM only (studies 1 and 2). A maximum *a posteriori* Bayesian (MAPB) analyses was performed on data from study 3 where there was one IM and one UM subjects. The model was adequate to fit these 2 subjects. Not enough data from genotypes other than EM were available to build a model with genotype as a covariate but the results of the MAPB analysis show that it would be something to explore in the future, pending data availability.

Figure 3: Predicted Metabolic Clearance of DM

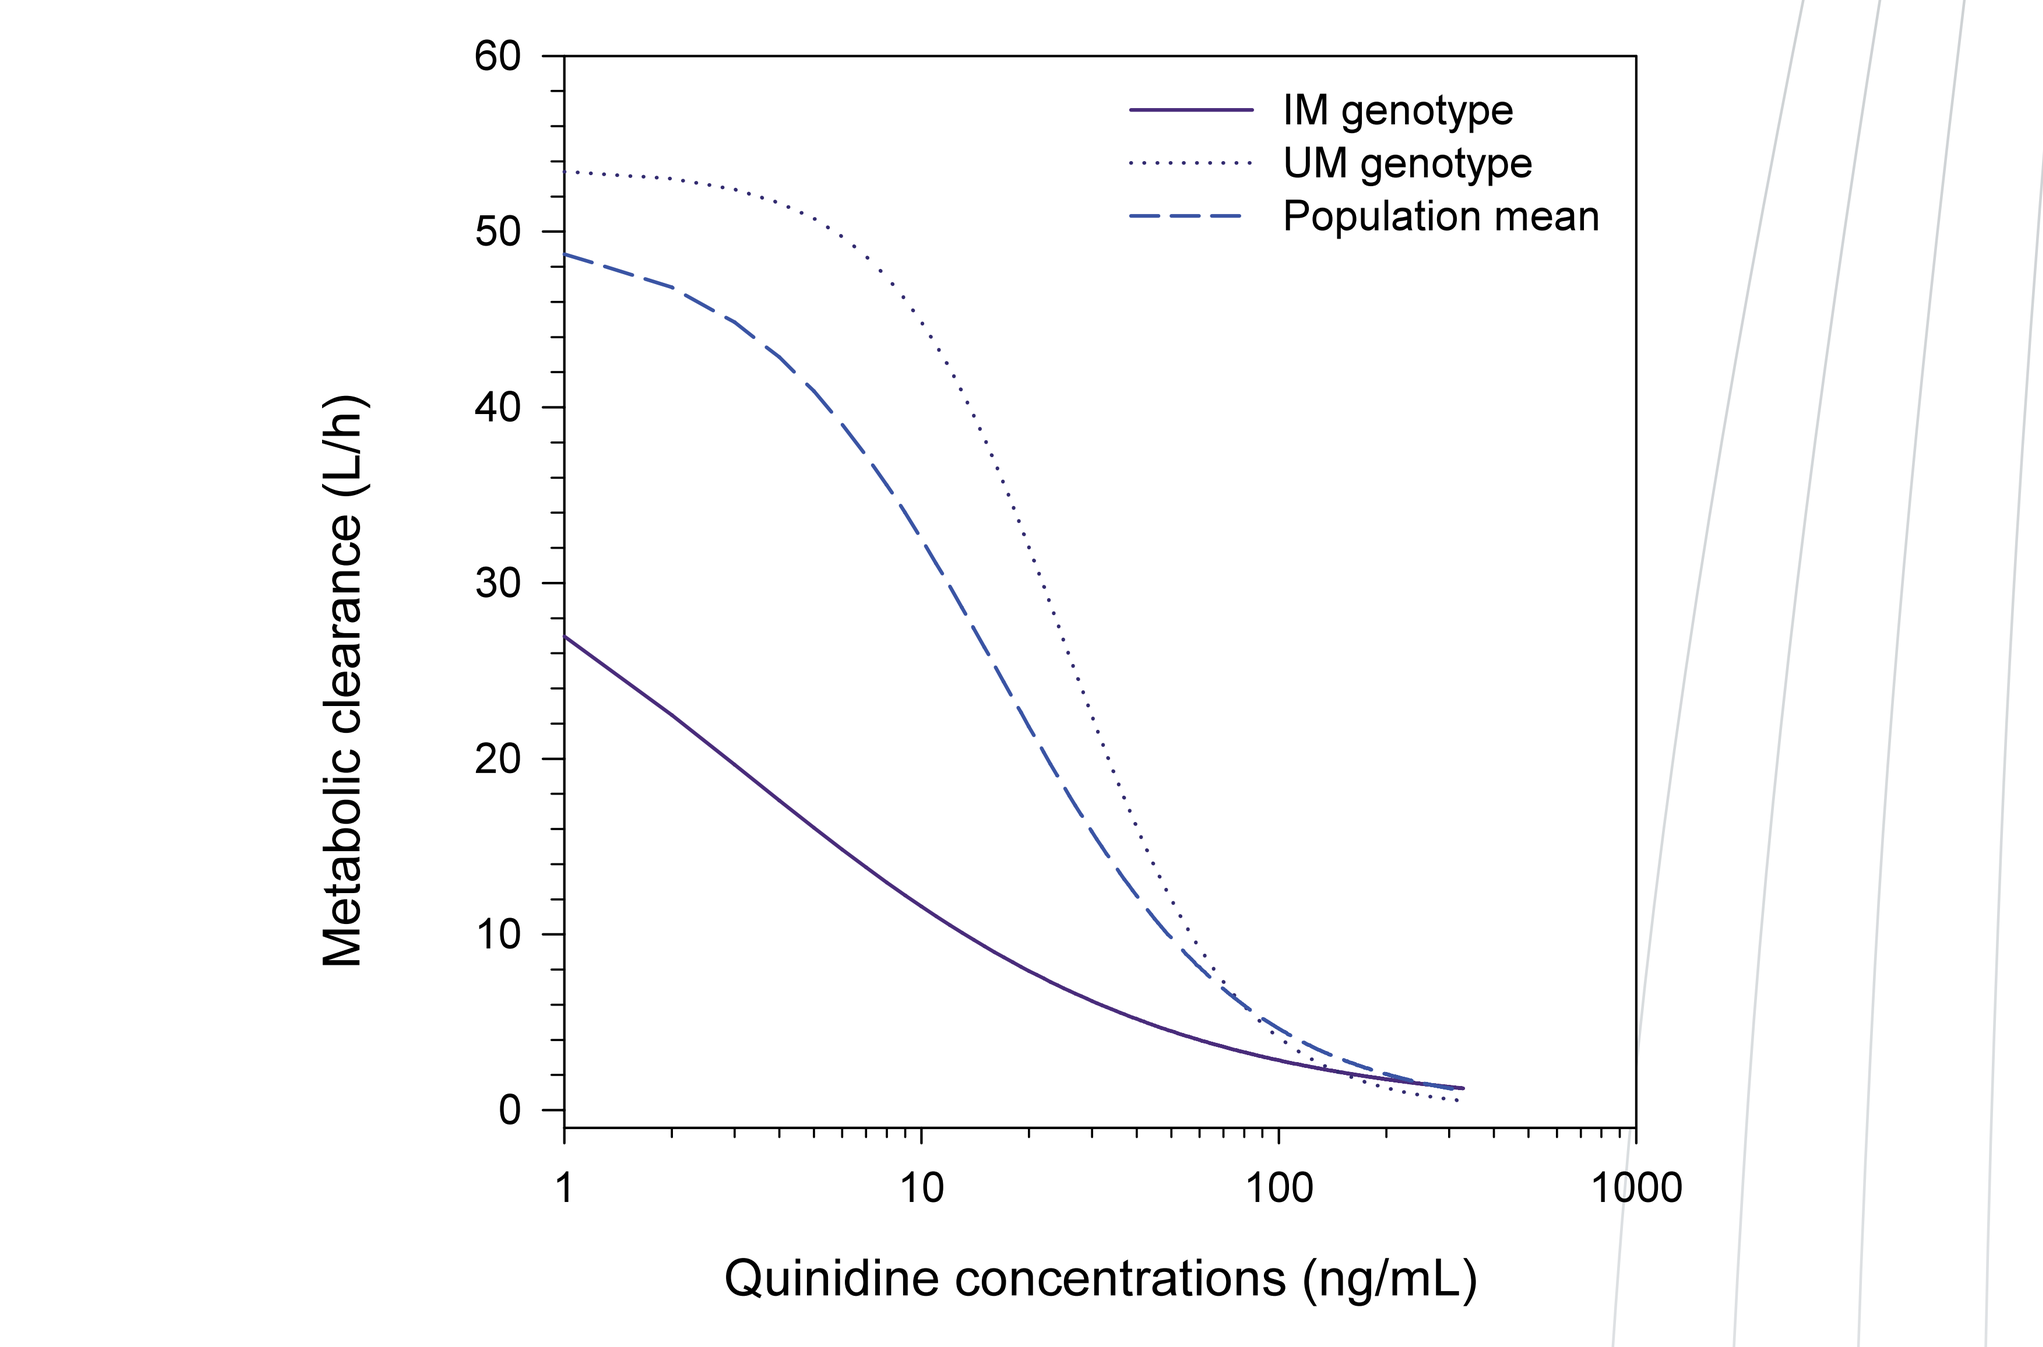
IM genotype
UM genotype
Population mean
Metabolic clearance (L/h)
Quinidine concentrations (ng/mL)

Figure 4: External Validation – Distribution of the Percentiles of Observed Concentrations

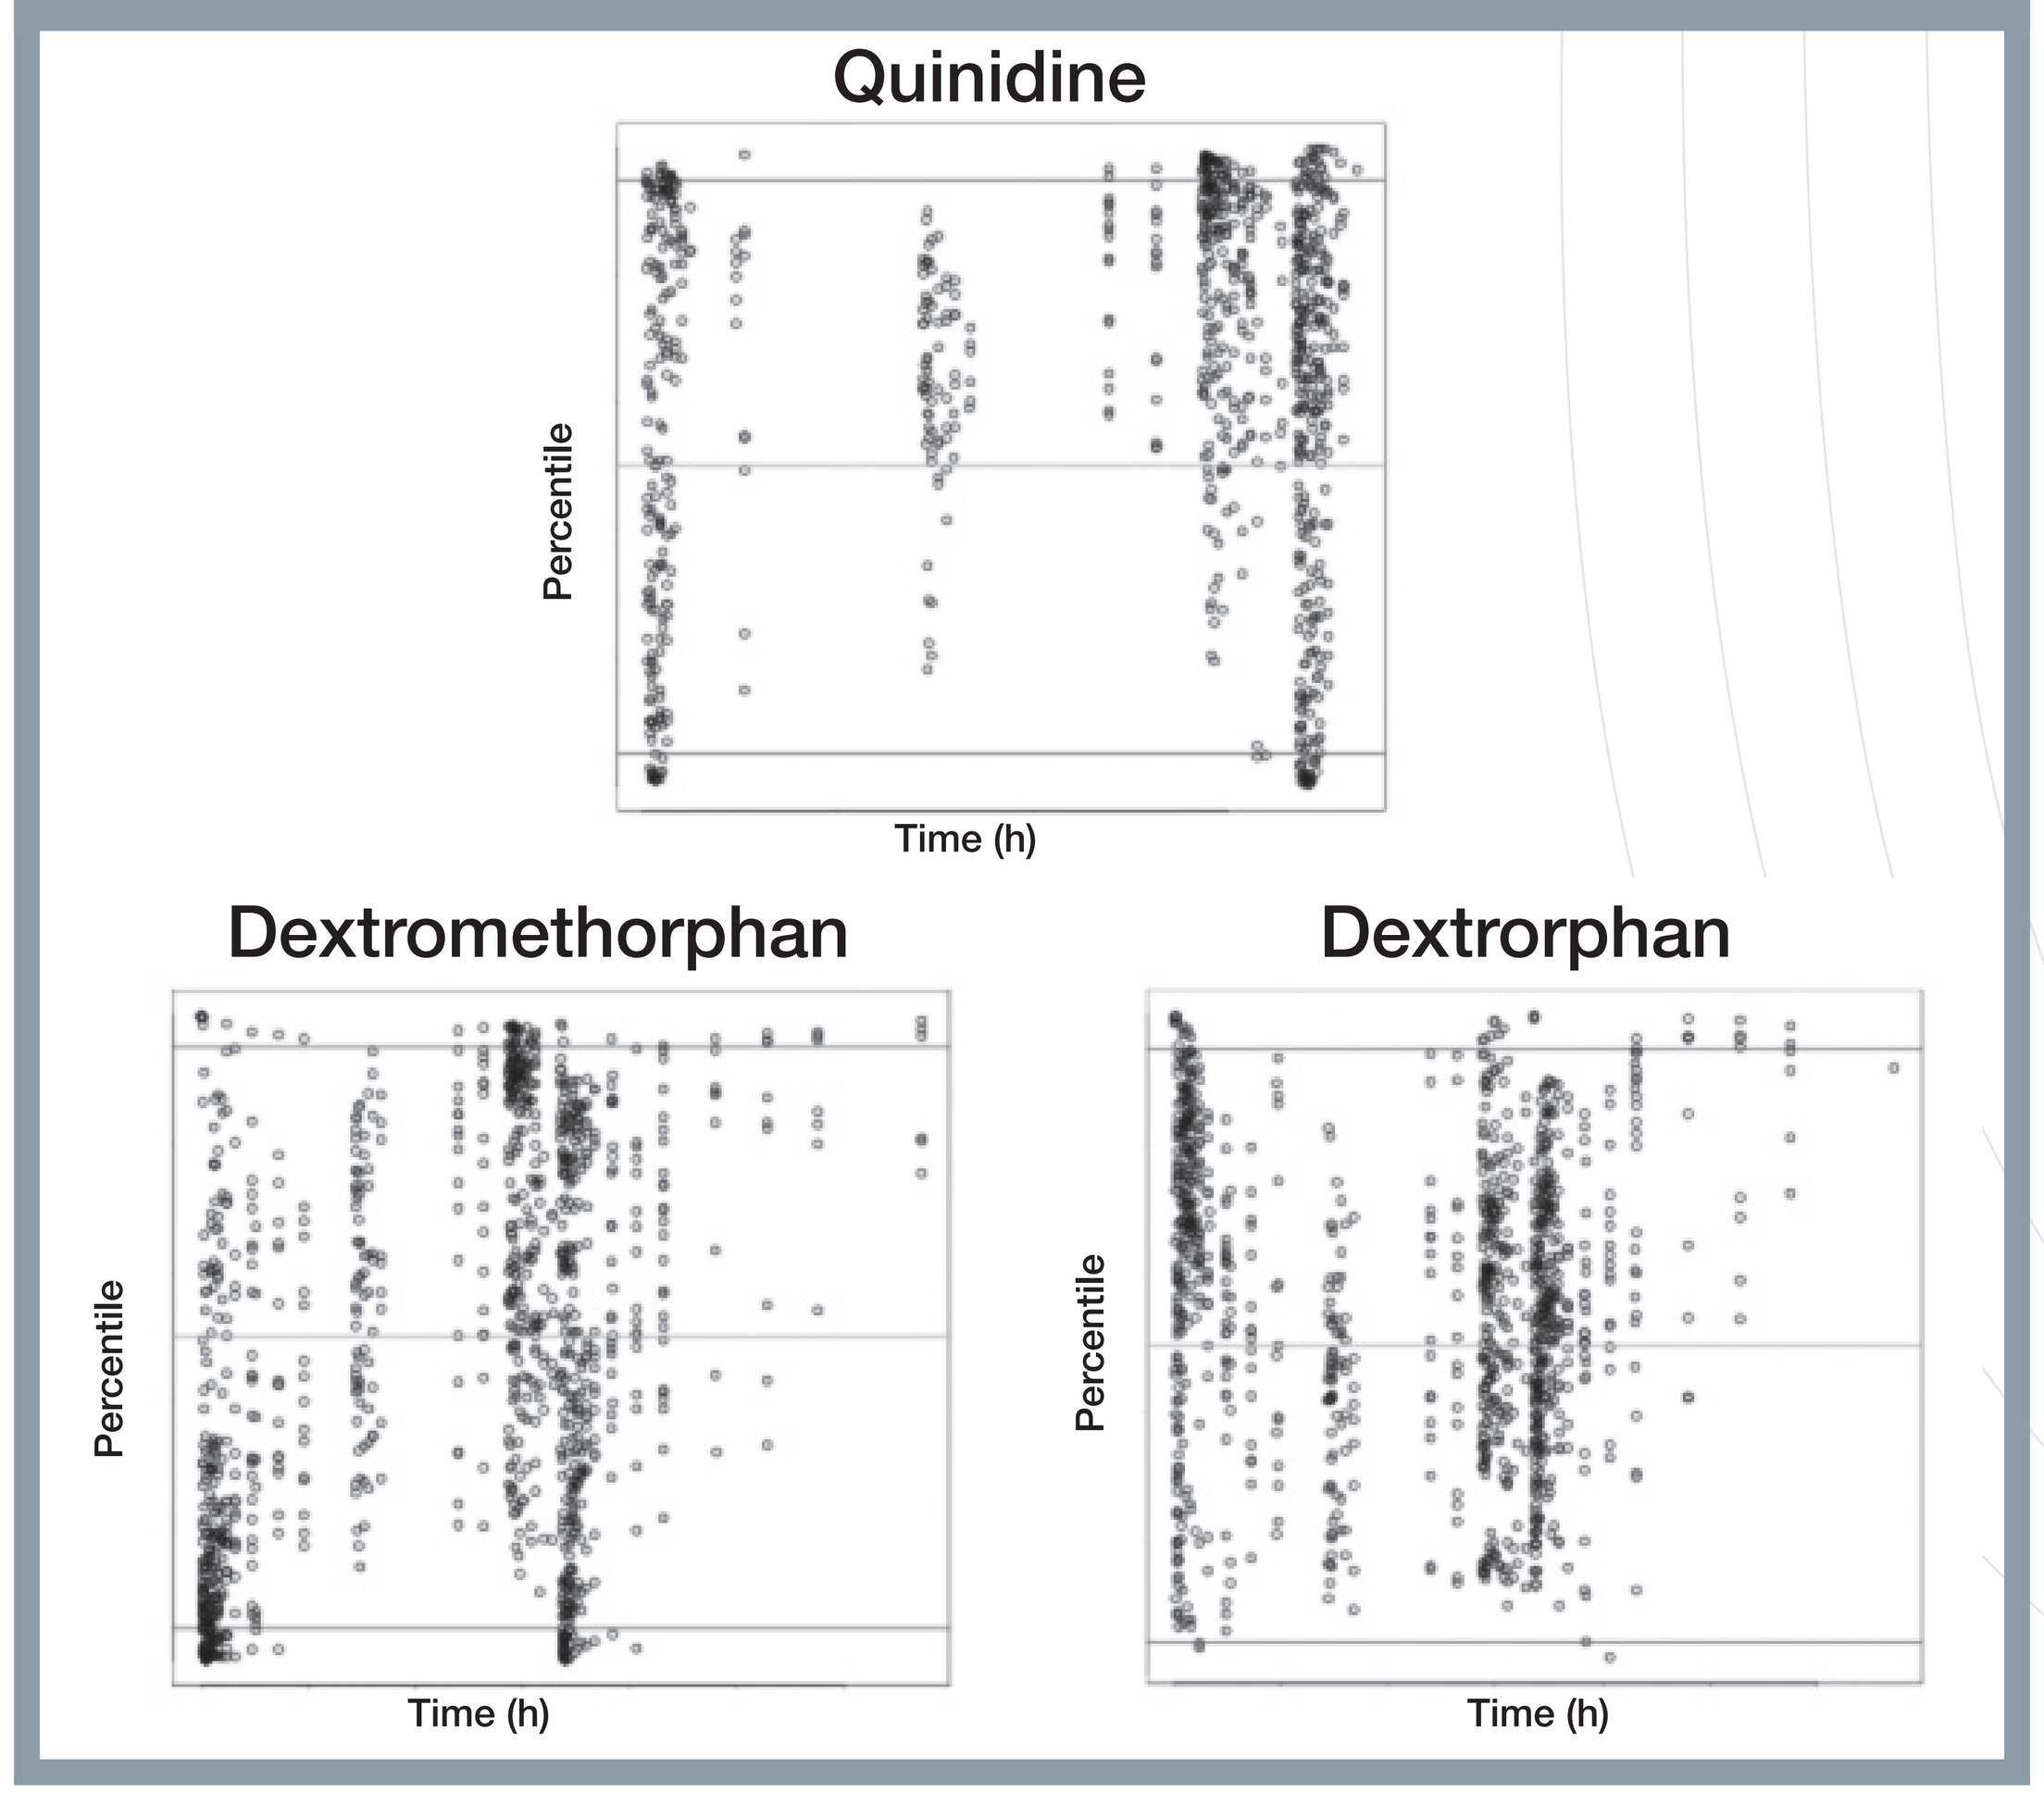
Quinidine
Dextromethorphan
Dextrorphan
Percentile
Time (h)

CONCLUSION

The PK of Q, DM and DX are well described by the population PK model developed. None of the available covariates were considered significantly correlated with any of the PK parameters. The external validation results show that the model predicts well the plasma PK of Q, DM and DX and can be used for the MAPB analysis of other sets of data or to predict the outcome of different dosing regimens for future clinical use.

REFERENCE

1. Wang D, Standardized Visual Predictive Check in Model Evaluation - Methodology and Applications. Downloaded from http://www.page-meeting.org/pdf_assets/4050-Standardized%20Visual%20Predictive%20Check%20in%20Model%2Evaluation%20-%20PAGE2009%20submit.pdf
2. D'Argenio, D.Z., A. Schumitzky and X. Wang. ADAPT 5 User's Guide: Pharmacokinetic/Pharmacodynamic Systems Analysis Software. BiomedicalSimulations Resource, Los Angeles, 2009.

www.celerion.com